010

Accidental Intra–Blood Vessel Auto-Injection of Adrenaline (Epinephrine) Rapidly Increases Pharmacokinetic and Hemodynamic Parameters

Motohiro Ebisawa, MD, PhD[1]; Michael A. Kaliner, MD[2]; Richard Lowenthal, MS, MBA[3]; Sarina Tanimoto, MD, PhD[3]
[1]Clinical Research Center for Allergy and Rheumatology, National Hospital Organization Sagamihara National Hospital, Sagamihara, Japan;
[2]Institute for Asthma & Allergy, Bethesda, MD, USA; [3]ARS Pharmaceuticals, Inc., San Diego, CA, USA

RATIONALE

- Many anaphylactic reactions occur outside of the hospital due to environmental allergen exposure (eg, insect bites, food, etc)
- Although adrenaline (epinephrine) auto-injectors are currently the primary product used for administration of adrenaline by patients and caregivers in a community setting, unintentional intra–blood vessel administration can occur, presenting a significant risk to patients, especially those with cardiovascular comorbidities
- Product labeling includes a warning that accidental intra–blood vessel injection of adrenaline may result in cerebral hemorrhage due to a sharp increase in blood pressure (BP) and notes cardiovascular reactions as potential adverse reactions, particularly in patients with heart disease
- We present the first documented case of accidental intra–blood vessel administration of adrenaline with an auto-injector supported by detailed pharmacokinetic (PK) and hemodynamic data

METHODS

- Japanese participants (n = 36) with seasonal allergies, aged 20 to 55 years, were randomized to receive adrenaline via the intranasal (1.0 mg) or intramuscular (0.3 mg via either needle and syringe or EpiPen) route of administration
- Blood samples for PK analysis were collected before dosing and at appropriate postdose intervals
- Noncompartmental methods were utilized in PK analysis of plasma–time data
- The Shinanozaka Clinic Clinical Trial Review Committee approved the study protocol; written informed consent was required from all participants

RESULTS

- A 55-year-old Japanese female experienced palpitations 1 minute after EpiPen adrenaline auto-injection
- BP increased from 118/79 mmHg to 221/128 mmHg 4 minutes after the injection (**Figure 1**)
 - A baseline pulse rate (PR) of 56 bpm increased to 71 bpm after 4 minutes and to 73 bpm after 5 minutes
 - For comparison, mean maximum increases in systolic BP, diastolic BP, and PR across all participants (n = 36) were 12.0 mmHg, 2.8 mmHg, and 16.3 bpm, respectively
- Adrenaline plasma levels reached 4370 pg/mL and 4390 pg/mL at 2 and 4 minutes after the injection, respectively (**Figure 2**)
 - The geometric mean maximum concentration (C_{max}) of all participants (n = 36) was 729 pg/mL
 - When this participant was removed from the PK analysis, the geometric mean C_{max} was 692.2 pg/mL, indicating that the accidental intra–blood vessel administration led to a 6.3-fold increase in maximum plasma adrenaline levels (**Figure 2**)
- The participant had no reported cardiovascular comorbidities and recovered without complications

Figure 1. Increase in Blood Pressure Following Accidental Intra–Blood Vessel Administration Compared With Successful Intramuscular Administration

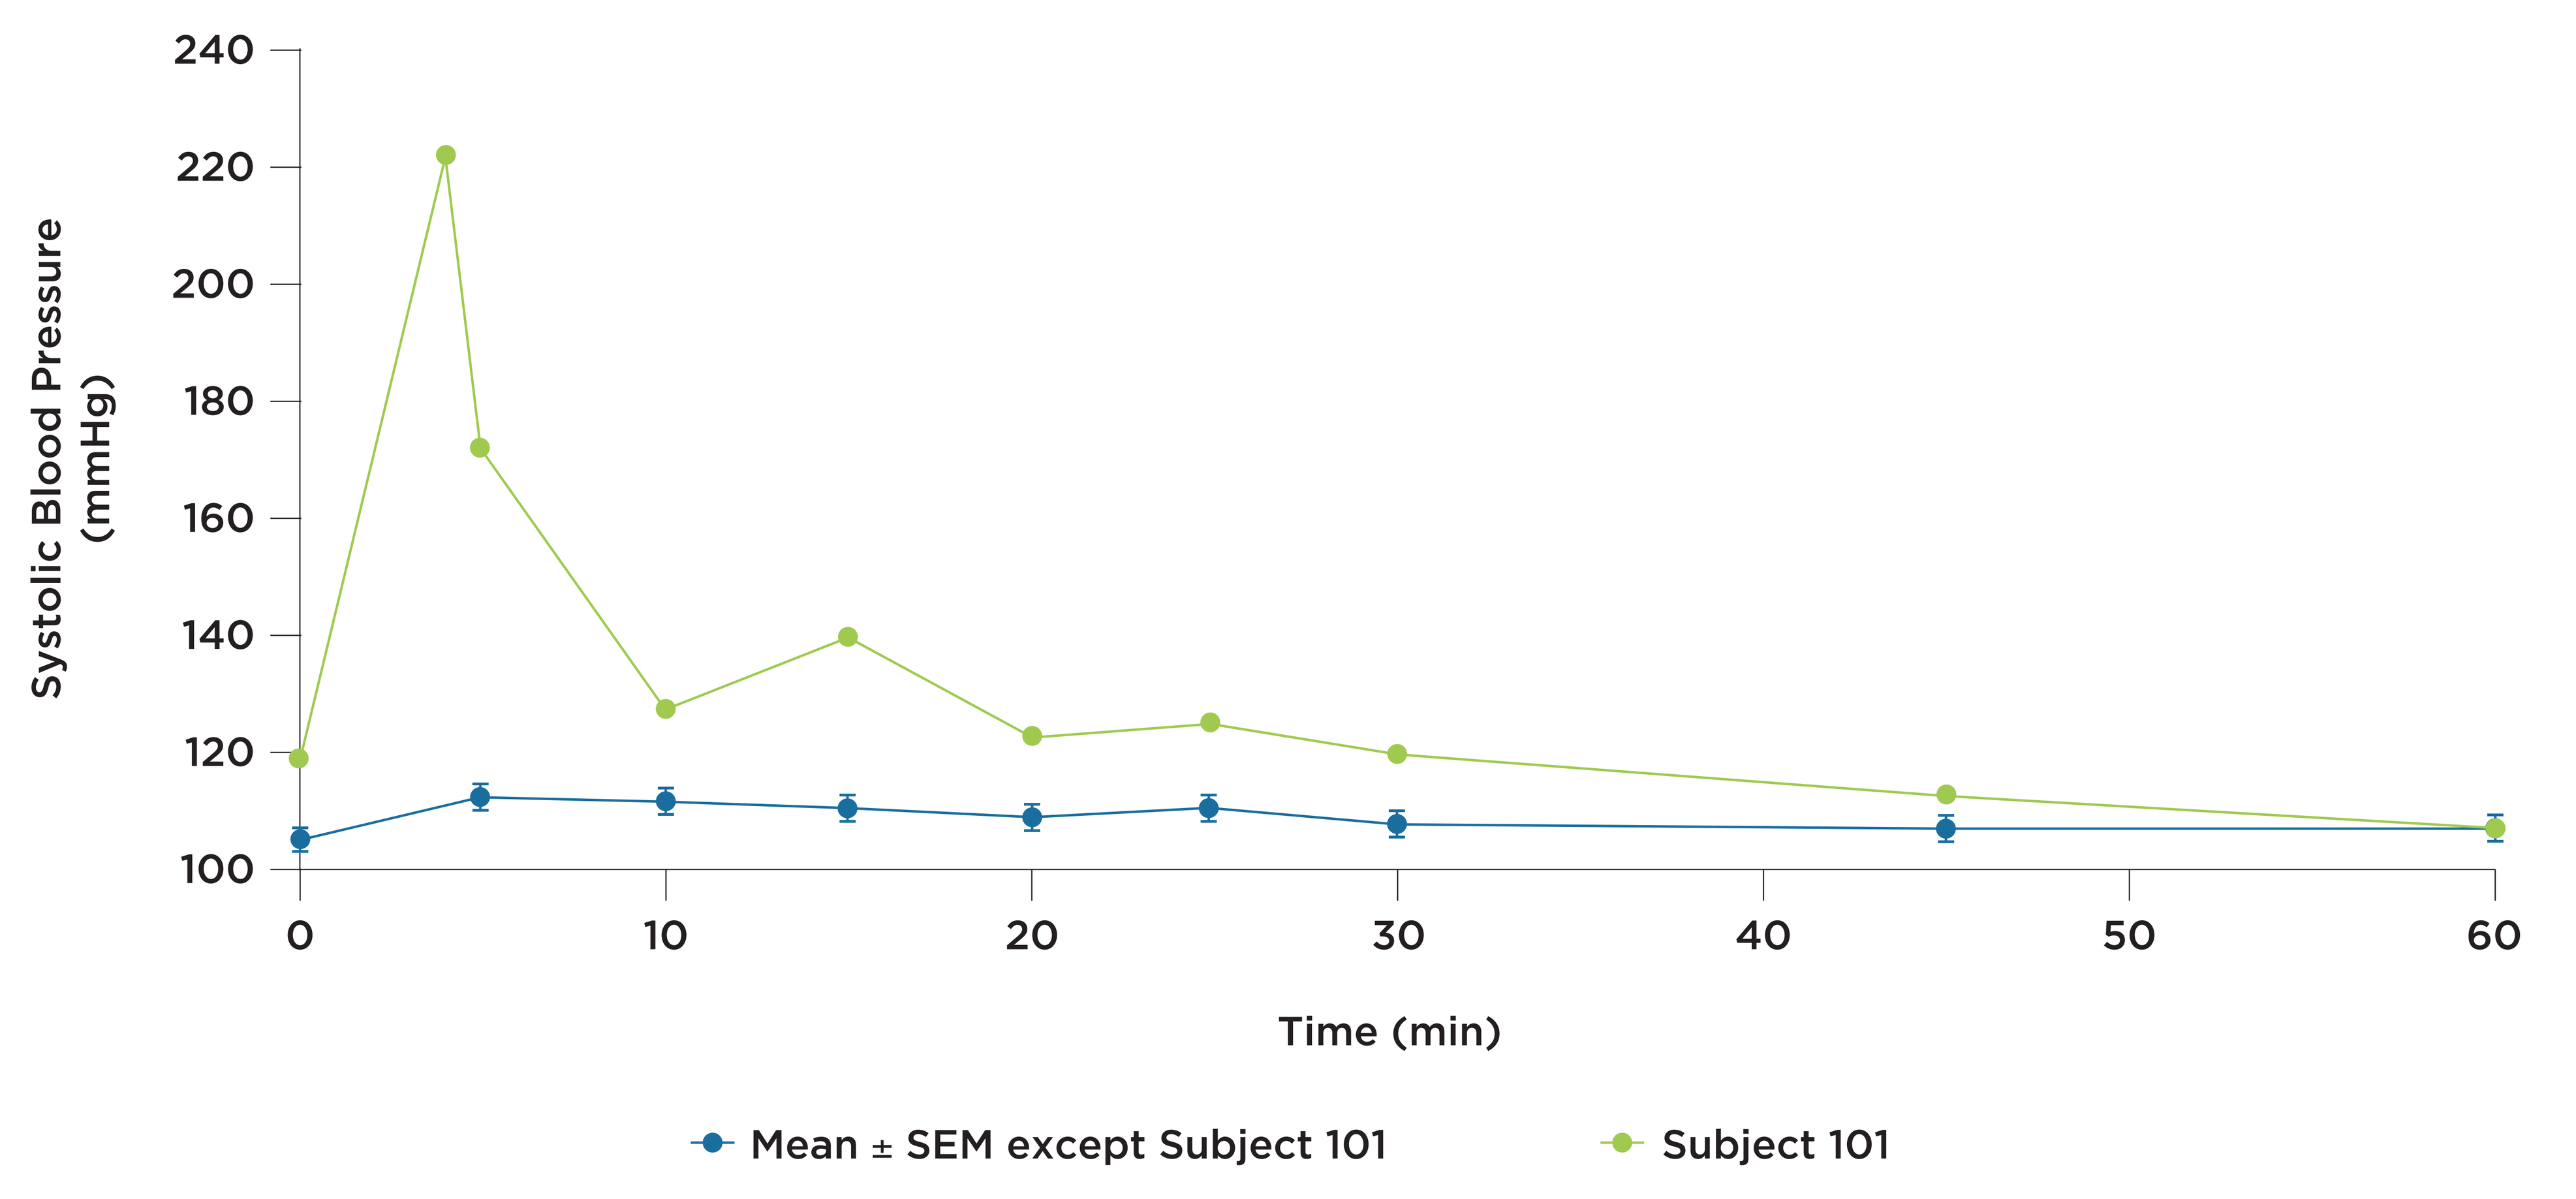
Systolic Blood Pressure (mmHg)
240
220
200
180
160
140
120
100
0
10
20
30
40
50
60
Time (min)
Mean ± SEM except Subject 101
Subject 101

SEM, standard error of the mean.

Figure 2. Increase in Plasma Adrenaline Levels Following Accidental Intra–Blood Vessel Administration Compared With Successful Intramuscular Administration

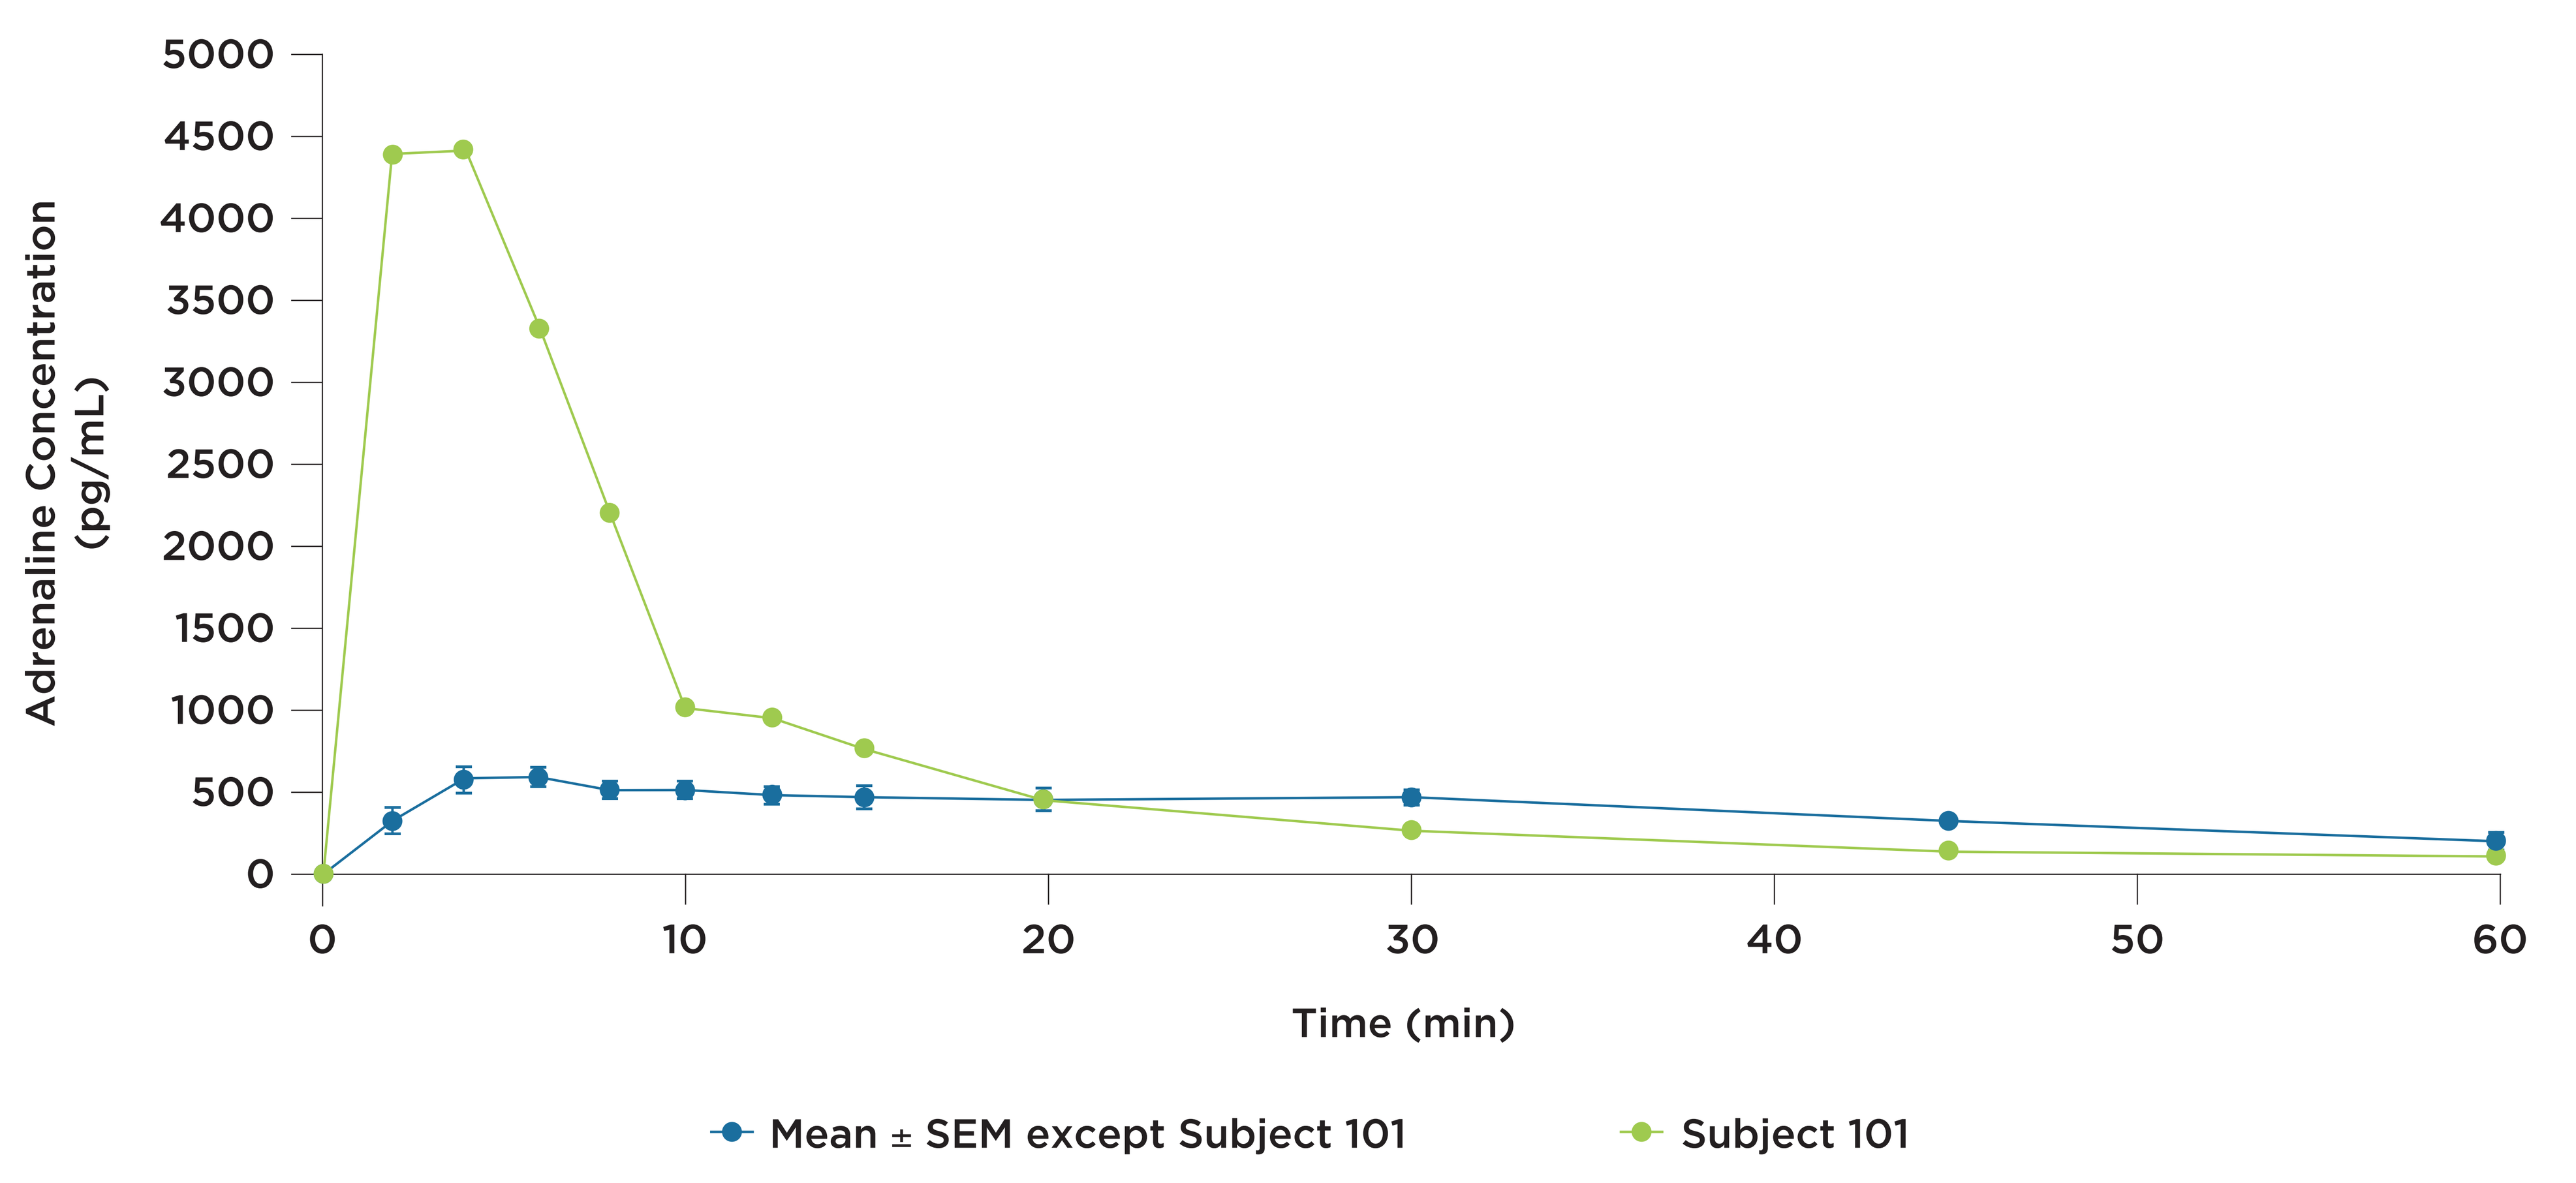
Adrenaline Concentration (pg/mL)
5000
4500
4000
3500
3000
2500
2000
1500
1000
500
0
0
10
20
30
40
50
60
Time (min)
Mean ± SEM except Subject 101
Subject 101

SEM, standard error of the mean.

DISCUSSION

- Several adrenaline auto-injectors have been approval by the US Food and Drug Administration and European Medicines Agency for the treatment of anaphylaxis. Current labeling states that adrenaline should be injected intramuscularly or subcutaneously into the anterolateral aspect of the thigh[1-5]
 - During typical use of an EpiPen auto-injector, the tip of the needle can penetrate to a depth of 30 to 40 mm
 - There is a risk of intra–blood vessel injection, as skeletal muscle is highly vascularized, and depending on body size, large blood vessels may be located within the 30- to 40-mm reach of the tip of an auto-injector needle[6]
- Furthermore, injections performed by non-expert users during an emergency may contribute to an increased risk of intra–blood vessel injection

CONCLUSIONS

- To our knowledge, this is the first report of PK and hemodynamic outcomes of intra–blood vessel adrenaline injection in a clinical setting
- Intra–blood vessel injection of adrenaline resulted in peak plasma levels >6 times higher than normal, leading to palpitations and sharp increases in BP and PR
- While there are warnings in the product labeling, this case shows that there is a very real risk of accidental intra–blood vessel injection associated with out-of-hospital adrenaline injection

REFERENCES

1. Song TT, Lieberman P. *Curr Opin Allergy Clin Immunol*. 2016;16:361-365.
2. Schwirtz A, Seeger H. *J Asthma Allergy*. 2012;5:39-49.
3. Ferguson JW, et al. *J Allergy Clin Immunol*. 2008;121:S25.
4. Song TT, et al. *Ann Allergy Asthma Immunol*. 2013;111:143-145.
5. EpiPen. Package insert. Meridian Medical Technologies, Inc.; 2016.
6. Nakajima Y, et al. *Hum Vaccin Immunother*. 2020;16:189-196.

Presented at the Annual Meeting of the American Academy of Allergy, Asthma and Immunology; February 25-28, 2022; Phoenix, AZ

Copyright © 2022. All rights reserved.