PHARMACOKINETICS/PHARMACODYNAMICS AFTER ADMINISTRATION OF ARS-1 (*neffy*® NASAL SPRAY), EPINEPHRINE AUTO-INJECTOR, AND MANUAL INTRAMUSCULAR INJECTION

A Nowak-Wegrzyn[1], R Lowenthal[2], S Tanimoto[2]

[1]NYU Grossman School of Medicine, New York, NY, USA; [2]ARS Pharmaceuticals, San Diego, CA, USA

RATIONALE

Epinephrine is considered the first-line treatment for severe allergic reactions and anaphylaxis. Epinephrine auto-injectors (EAIs) are the most frequently used products for out-of-hospital treatment; however, they are considered inconvenient and cumbersome.

Up to 83% of patients/caregivers reporting failing to administer or delaying use of EAIs, even during a severe allergic reaction.[1] Additionally, the Asthma and Allergy Foundation of America has reported that 72% of parents did not administer epinephrine to their child, even when they knew the child was experiencing a severe allergic reaction.[2]

Delayed treatment, defined as treatment after presentation to the emergency department, is associated with a significantly increased risk of hospitalization (odds ratio 4.0 [95% CI, 0.12-0.49]).[3]

A survey of 200 epinephrine users (100 patients and 100 caregivers) demonstrated that the mean time to dose was significantly higher in patients and caregivers who self-reported delaying treatment or were hesitant to treat (12.6 minutes vs 6.1 minutes, p<0.01) (Figure 1).

There were several factors that were significantly associated with delayed dosing, including the requirement to go to ER, uncertainty about the symptom being warranted for injection, potential side effects, fear to use (p<0.01), pain, and size of device (p<0.05) (Figure 2). These results suggest that an alternative device may decrease hesitancy among patients and caregivers and reduce the time to dose.

ARS Pharmaceuticals, Inc is developing *neffy*, an intranasal (IN) epinephrine spray that is a needle-free alternative epinephrine delivery device for the emergency treatment of (Type I) allergic reactions, including anaphylaxis.

neffy is expected to have significant clinical benefit by reducing apprehension and delay in dosing, reducing dosing errors and making it easier to carry the product at all times. *neffy* is anticipated to have pharmacokinetic, pharmacodynamic, and safety profiles that are within the range of currently approved epinephrine injection products.

Figure 1: Time to Use by Hesitation and Delay (Yes/No)

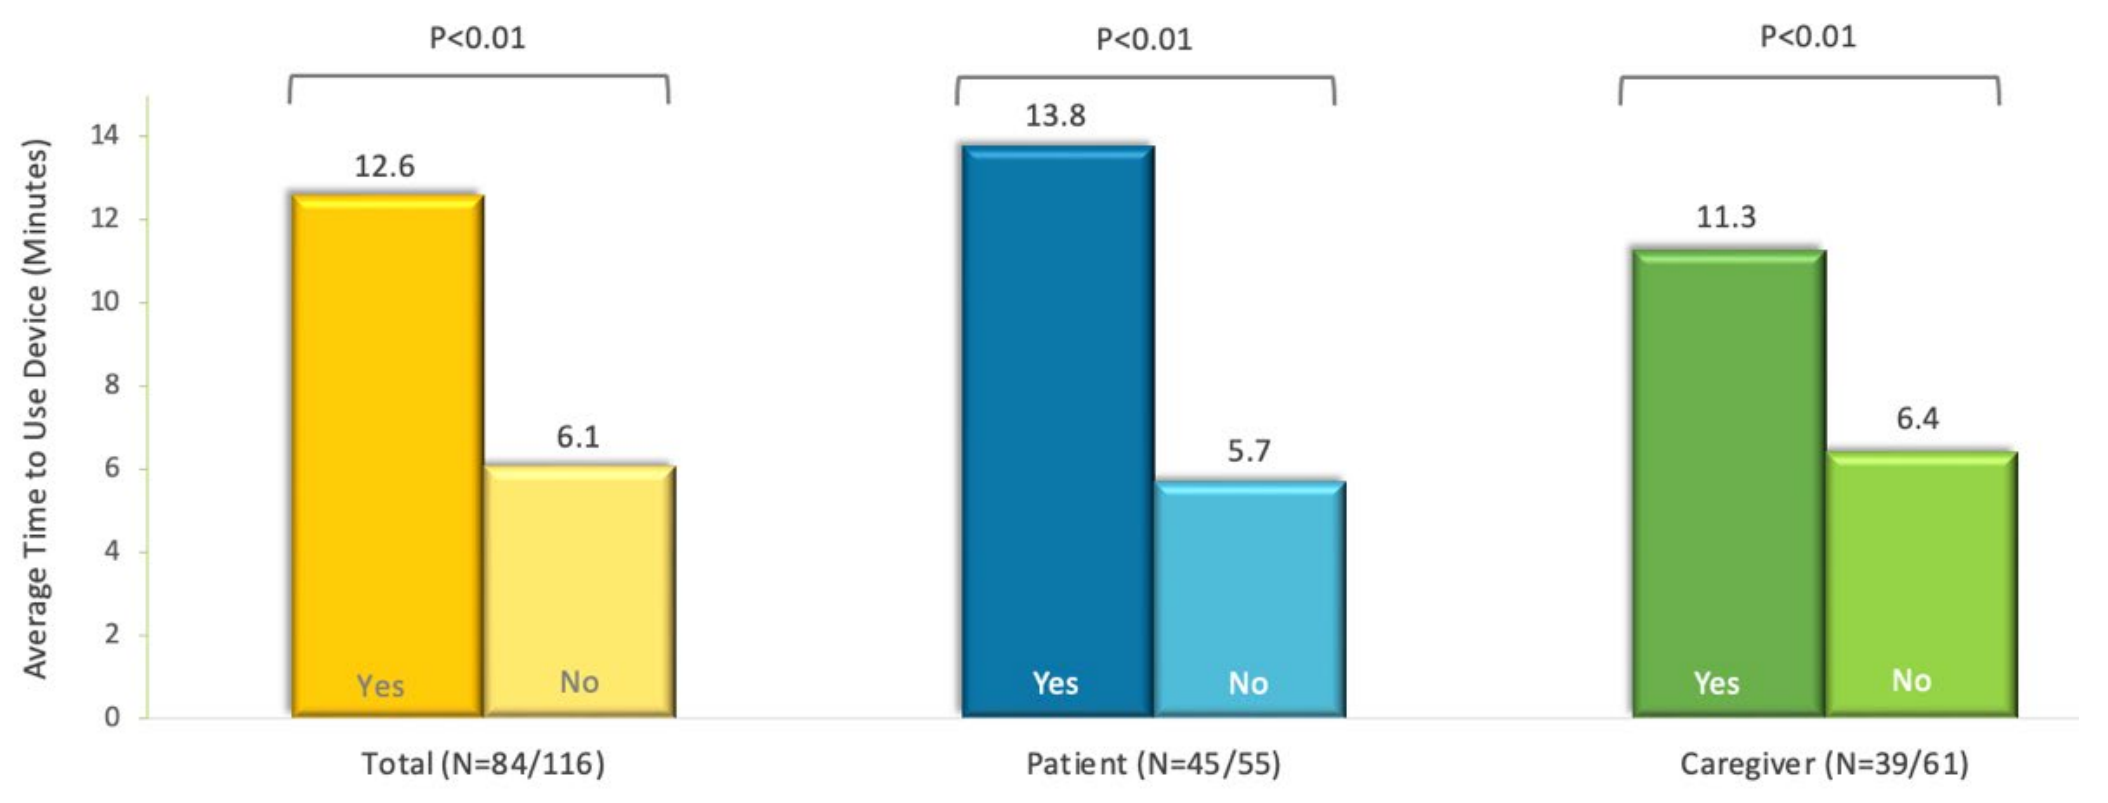
Average Time to Use Device (Minutes)
0
2
4
6
8
10
12
14
P<0.01
12.6
6.1
Yes
No
Total (N=84/116)
P<0.01
13.8
5.7
Yes
No
Patient (N=45/55)
P<0.01
11.3
6.4
Yes
No
Caregiver (N=39/61)

Figure 2: Reason for Delay or Hesitation Using Device with Statistical Difference (P<0.01 or P<0.05)

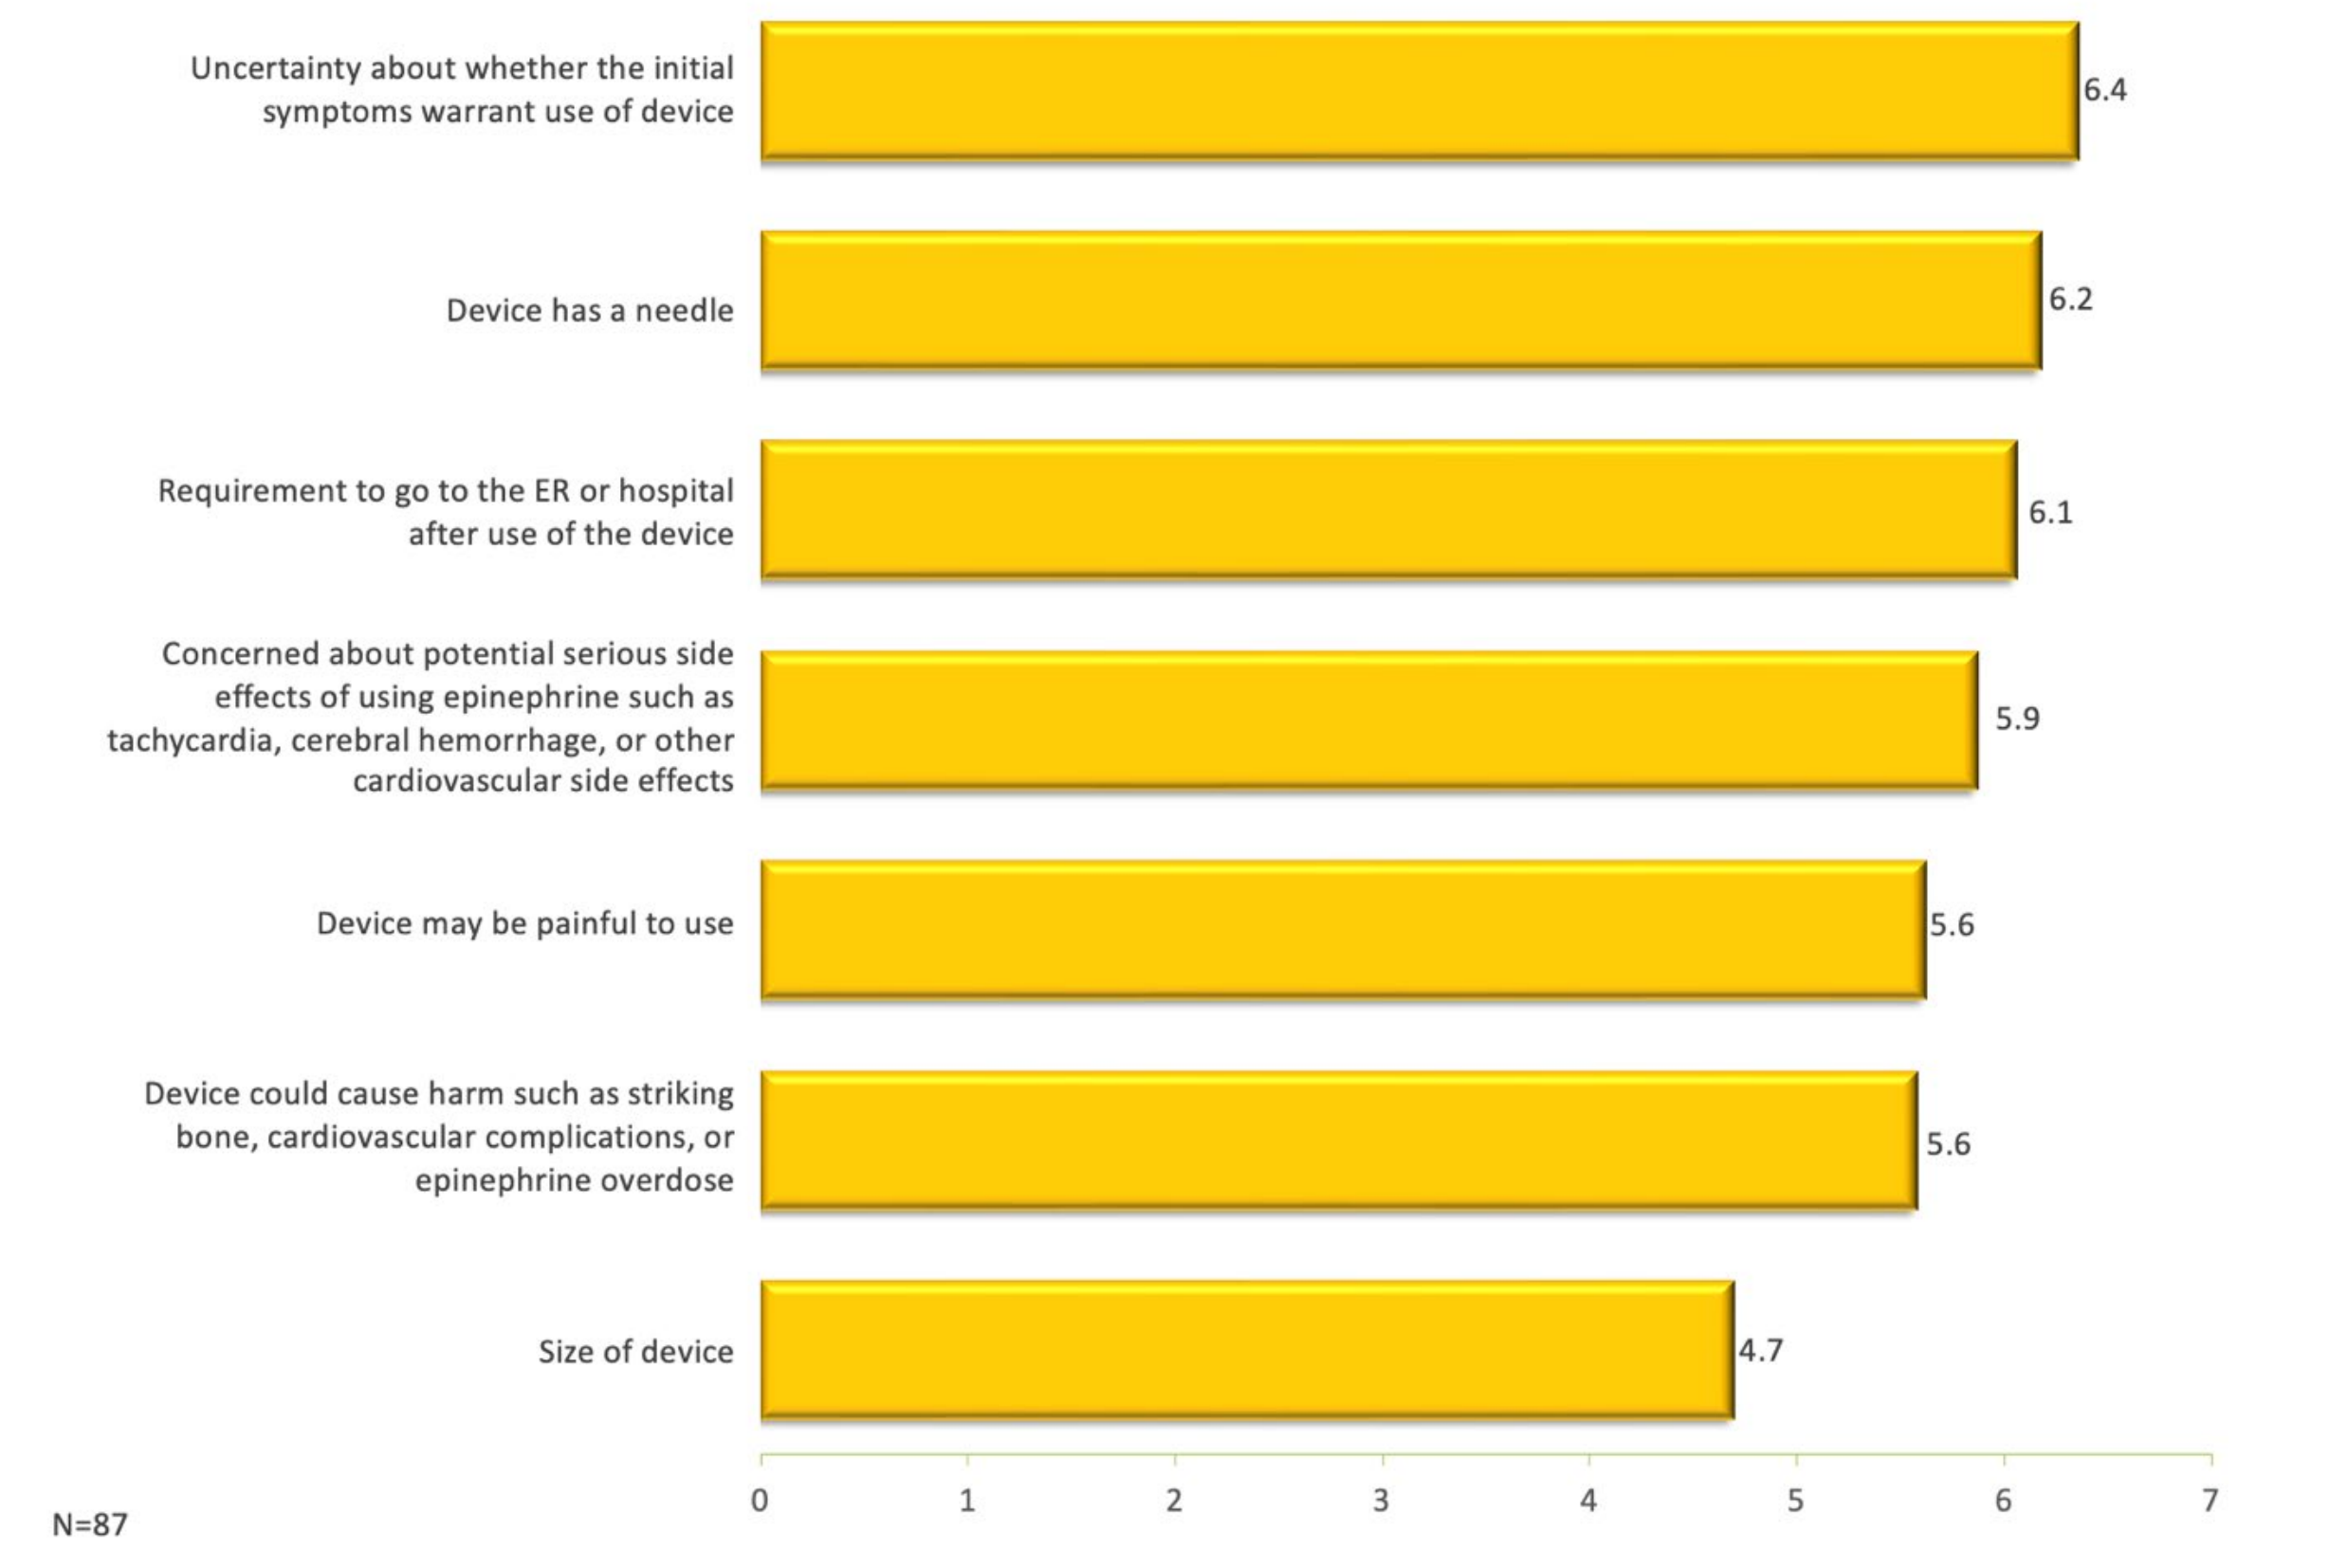

Uncertainty about whether the initial symptoms warrant use of device
6.4
Device has a needle
6.2
Requirement to go to the ER or hospital after use of the device
6.1
Concerned about potential serious side effects of using epinephrine such as tachycardia, cerebral hemorrhage, or other cardiovascular side effects
5.9
Device may be painful to use
5.6
Device could cause harm such as striking bone, cardiovascular complications, or epinephrine overdose
5.6
Size of device
4.7
0
1
2
3
4
5
6
7
N=87

METHODS

EPI 15 was one of a series of studies conducted to evaluate the pharmacokinetics and pharmacodynamics of *neffy*. This pivotal study was a Phase 1, six-treatment, six-period, crossover study in 59 healthy subjects. Each subject received:

- a single dose of *neffy* 2.0 mg IN
- a single dose of EpiPen® 0.3 mg
- a single dose of Epinephrine manual 0.3 mg Intramuscular Injection (IM)

Pharmacokinetics (mean epinephrine concentrations and epinephrine pharmacokinetic parameters) and pharmacodynamics (systolic blood pressure [SBP], diastolic blood pressure [DBP], and heart rate [HR]) were evaluated for each treatment.

RESULTS

<u>Pharmacokinetic Results</u>

Mean epinephrine concentrations were highest for EpiPen until approximately 20 minutes post-dose. From 30 minutes to 360 minutes post-dose, *neffy* exhibited higher mean concentrations compared to EpiPen and Epinephrine IM.

Mean C_{max} values were highest after EpiPen (753 pg/mL), followed by *neffy* (481 pg/mL), and Epinephrine IM (339 pg/mL). The greatest total exposure was observed after *neffy* (43500 min*pg/mL), followed by EpiPen (31300 min*pg/mL), and Epinephrine IM (29300 min*pg/mL). Median t_{max} values were fastest following EpiPen (7.50 minutes), followed by *neffy* (30.0 minutes), and Epinephrine IM (45.0 minutes) (Figure 3 and Table 1).

Figure 3: Change from Baseline - Mean Epinephrine Concentration vs. Time Profiles

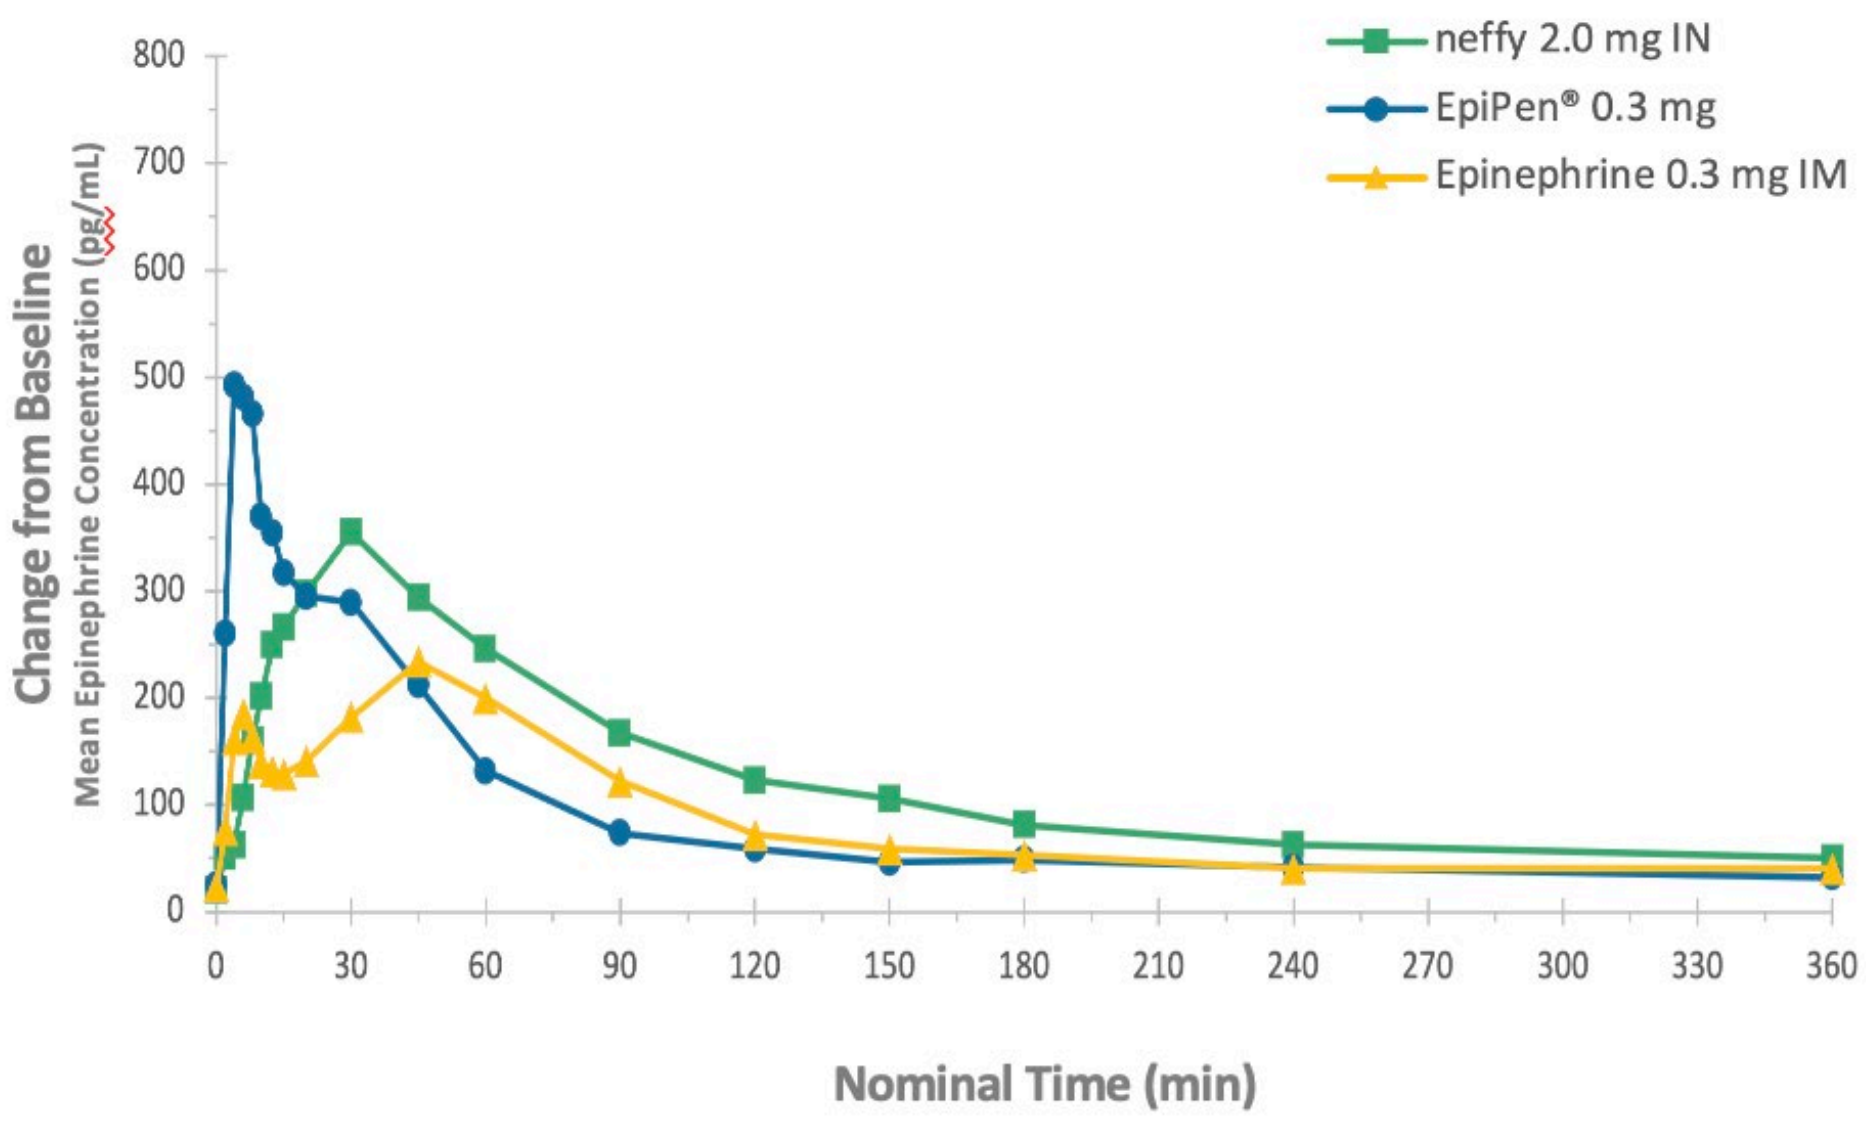

neffy 2.0 mg IN
EpiPen® 0.3 mg
Epinephrine 0.3 mg IM
Change from Baseline
Mean Epinephrine Concentration (pg/mL)
800
700
600
500
400
300
200
100
0
0
30
60
90
120
150
180
210
240
270
300
330
360
Nominal Time (min)

Table 1: Summary Statistics of Epinephrine Pharmacokinetic Parameters

Treatment	N	t_{max} (min) median (range)	C_{max} (pg/mL) mean (%CV)	AUC_{last} (min*pg/mL) mean (%CV)
Single Dose				
***neffy* 2.0 mg IN**	42	30.0 (6.00 – 150)	481 (76.0)	43500 (69.4)
EpiPen 0.3 mg	42	7.50 (2.00 – 45.0)	753 (65.6)	31300 (35.0)
Epinephrine 0.3 mg IM	42	45.0 (4.00 – 90.0)	339 (74.1)	29300 (41.7)

C_{max} = maximum plasma concentration; T_{max} = time to maximum plasma concentration,
AUC_{last} = area under the curve to the final time with a concentration equal to or greater than the lower limit of quantitation

Pharmacodynamic Results

Systolic Blood Pressure (Figure 4 and Table 2)

All treatments resulted in an increase from baseline SBP, with the greatest increase observed following *neffy*. EpiPen resulted in a smaller and more abrupt increase relative to *neffy* and only a minimum increase was observed after Epinephrine IM. Mean SBP E_{max} was higher following *neffy* relative to both EpiPen and Epinephrine IM.

Diastolic Blood Pressure (Figure 4 and Table 2)

neffy resulted in an initial increase from baseline DBP, followed by a decrease from baseline. Both EpiPen and Epinephrine IM resulted in an immediate decrease from baseline DBP. The decrease from baseline was markedly more pronounced following EpiPen and Epinephrine IM relative to *neffy*. Mean DBP E_{max} was significantly different following *neffy* relative to Epinephrine IM and EpiPen.

Heart Rate (Figure 4 and Table 2)

All treatments resulted in increases from baseline HR. There was a return towards baseline after both Epinephrine IM and EpiPen, while the elevation persisted throughout the 120 minutes following *neffy*. In general, E_{max} was significantly higher following *neffy* relative to EpiPen and Epinephrine IM.

Figure 4: Mean Change from Baseline Pharmacodynamics vs. Time Profiles

Systolic Blood Pressure (SBP)	Diastolic Blood Pressure (DBP)

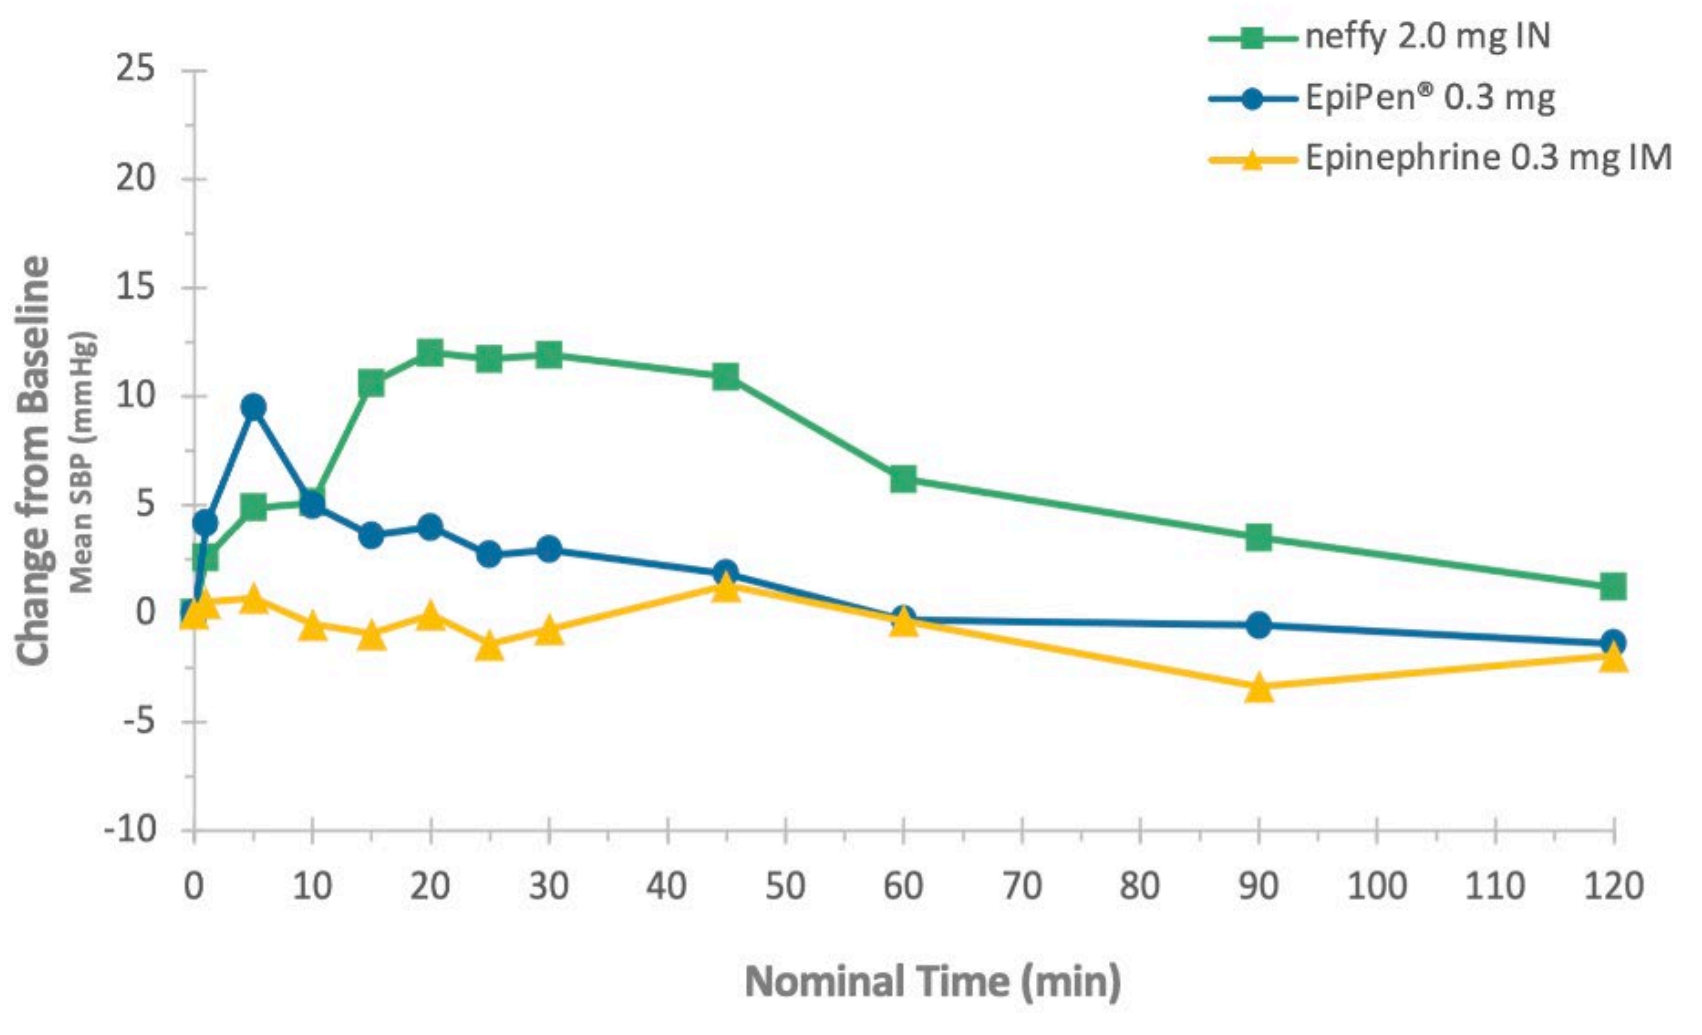

neffy 2.0 mg IN
EpiPen® 0.3 mg
Epinephrine 0.3 mg IM
Change from Baseline
Mean SBP (mmHg)
25
20
15
10
5
0
-5
-10
0 10 20 30 40 50 60 70 80 90 100 110 120
Nominal Time (min)

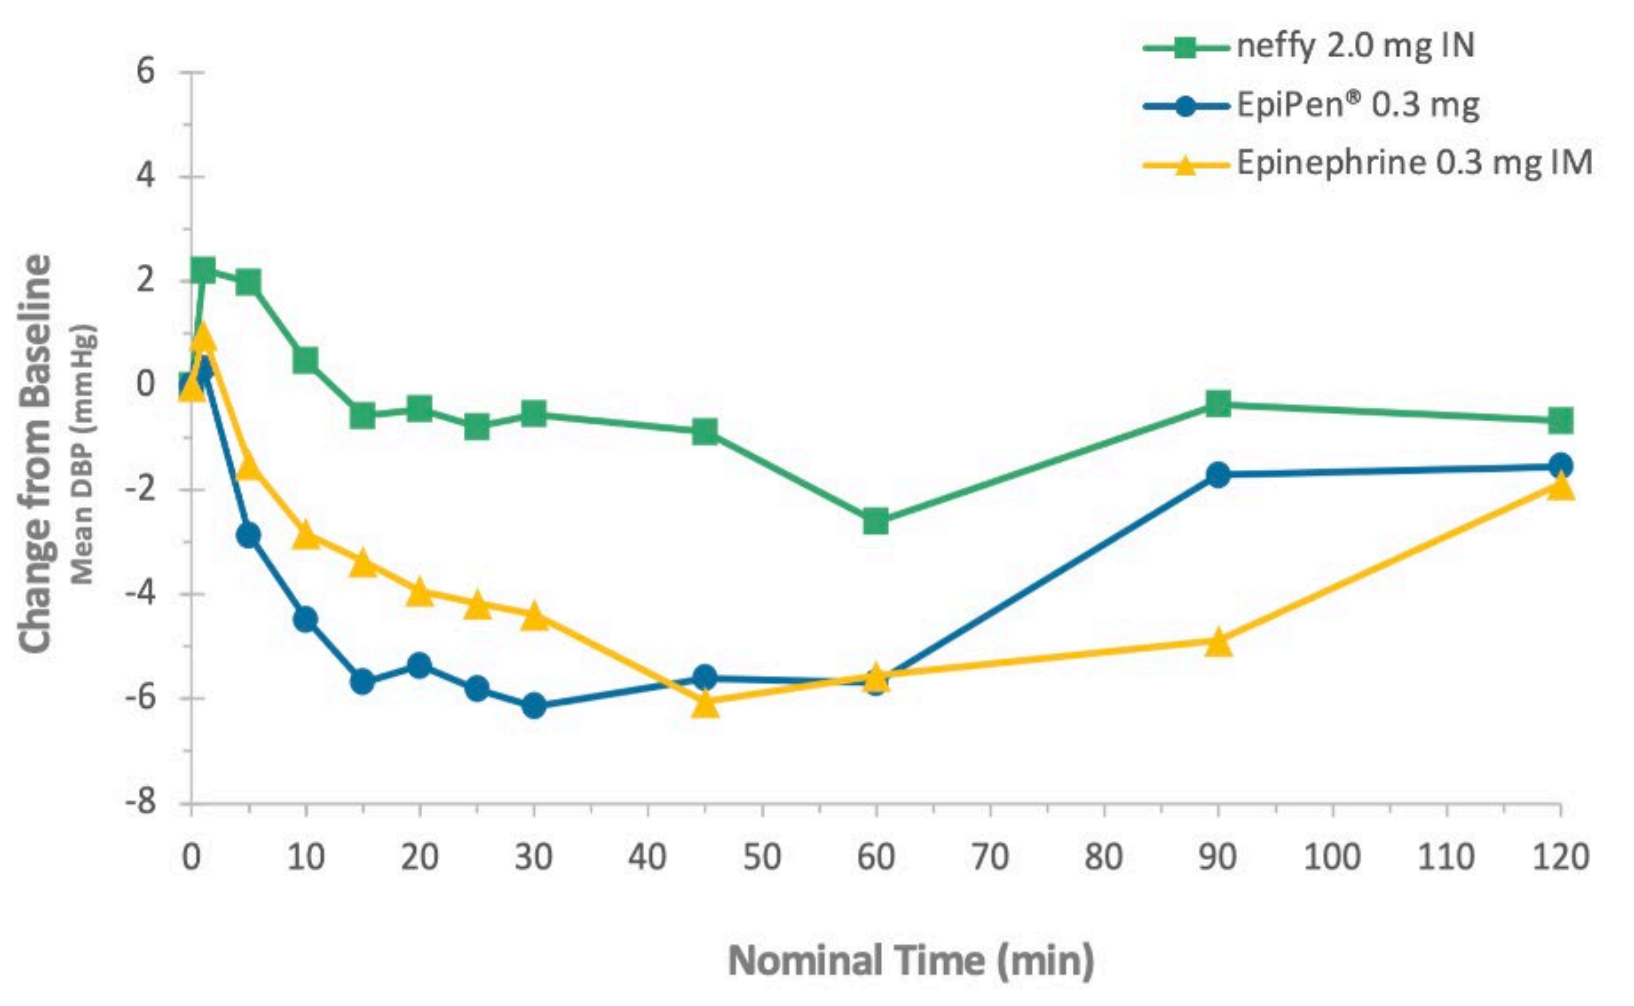

neffy 2.0 mg IN
EpiPen® 0.3 mg
Epinephrine 0.3 mg IM
Change from Baseline
Mean DBP (mmHg)
6
4
2
0
-2
-4
-6
-8
0 10 20 30 40 50 60 70 80 90 100 110 120
Nominal Time (min)

Heart Rate (HR)	

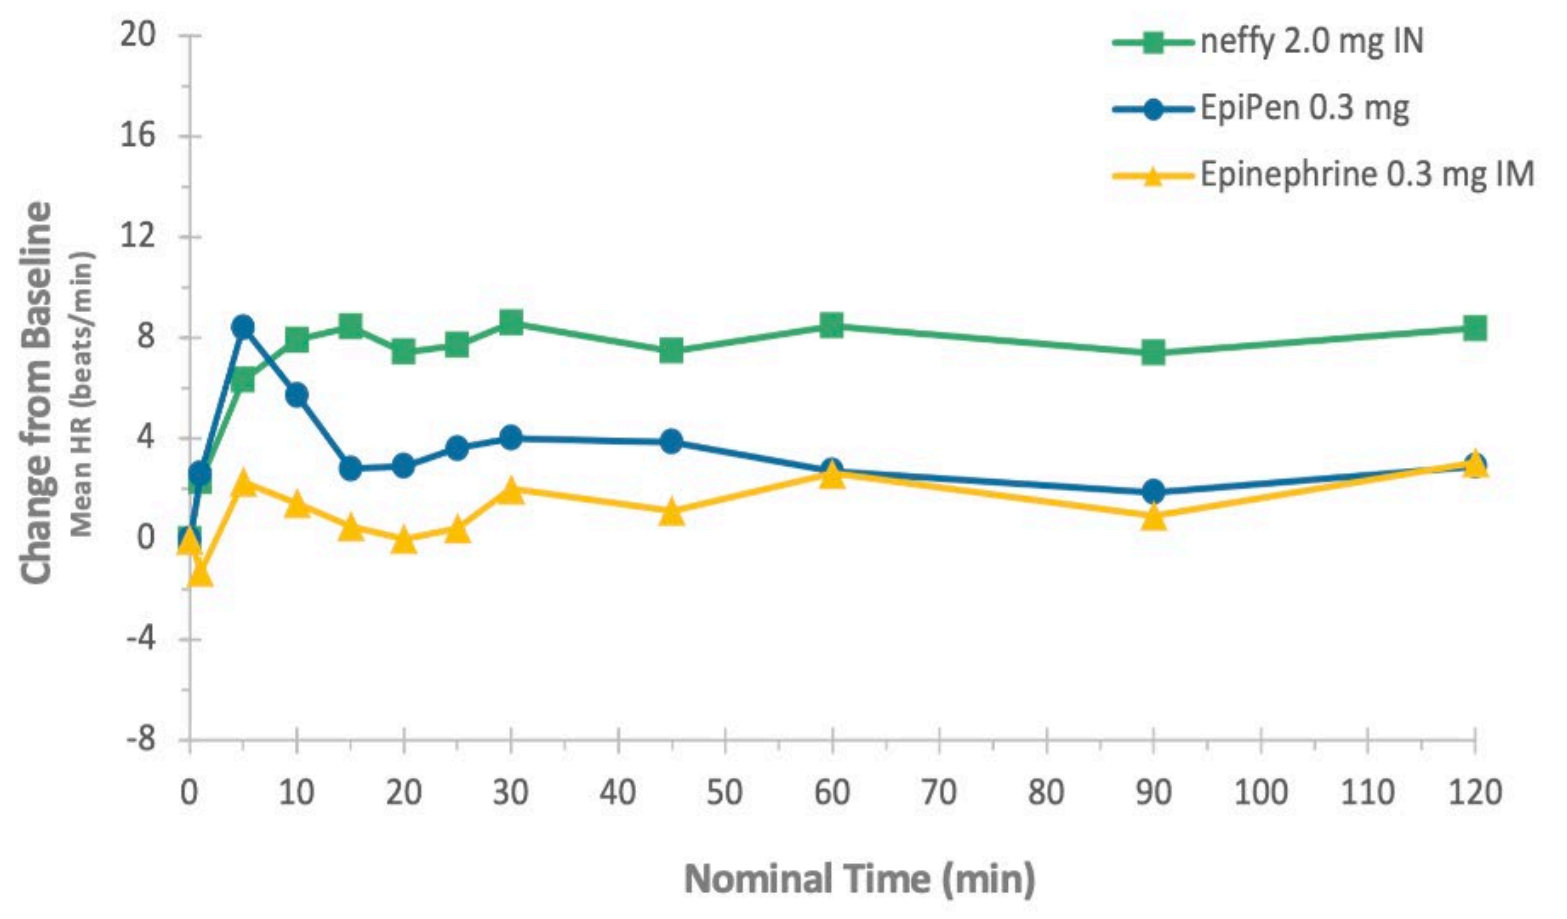

neffy 2.0 mg IN
EpiPen 0.3 mg
Epinephrine 0.3 mg IM
Change from Baseline
Mean HR (beats/min)
20
16
12
8
4
0
-4
-8
0 10 20 30 40 50 60 70 80 90 100 110 120
Nominal Time (min)

Table 2: Maximum Pharmacodynamic Effect (Change from Baseline) and Time to Maximum Pharmacodynamic Effect

Treatment	N	Mean E_{max} (CV)			Median T_{max} (min)		
		SBP (mmHg)	DBP (mmHg)	HR (bpm)	SBP	DBP	HR
Single Dose							
***neffy* 2.0 mg IN**	42	23.6 (64.8)	8.10 (64.3)	17.3 (62.7)	25.0 (1.00 – 116)	13.0 (1.00 – 117)	19.0 (1.00 – 116)
EpiPen 0.3 mg	42	18.2 (80.3)	5.62 (131)	12.3 (63.2)	9.0 (1.00 – 116)	10.0 (1.00 – 115)	10.0 (1.00 – 115)
Epinephrine 0.3 mg IM	42	11.9 (81.0)	5.48 (145)	9.71 (87.1)	22.5 (1.00 – 116)	9.00 (1.00 – 115)	27.0 (1.00 – 117)

E_{max} = maximum effect; TE_{max} = time to maximum effect;
SBP = systolic blood pressure; DBP = diastolic blood pressure; PR = pulse rate

Safety Results

The study treatments were well tolerated, and all treatment emergent adverse events were considered mild. The maximum increase in SBP was 90 mmHg, which was within the range of the currently approved products.

CONSLUSIONS

neffy 2.0 mg IN has a pharmacokinetic profile that is within the range of currently approved injection products (Epinephrine 0.3 mg IM and EpiPen 0.3 mg).

The *neffy* 2.0 mg IN Pharmacodynamic profile is comparable to EpiPen 0.3 mg and is comparable to or better than Epinephrine 0.3 mg IM, suggesting that *neffy* is expected to be at least as efficacious as EpiPen.

Despite having a lower C_{max} relative to EpiPen, *neffy* resulted in more pronounced increases in SBP, DBP, and PR. One of the mechanisms by which this occurs likely involves *neffy's* ability to bypass the powerful β_2-mediated vasodilation in the skeletal muscle.[4] In contrast to intranasal administration, intramuscular injection in the thigh directly exposes skeletal muscle to the full dose of epinephrine, resulting in the activation of the β_2 receptors that are abundant in the skeletal muscle. This β_2 activation results in vasodilation and a subsequent decrease in peripheral vascular resistance, ultimately resulting in a rapid decrease in DBP.[5]

The pharmacokinetic and pharmacodynamic results of this study demonstrate that *neffy* has the potential to be a safe, effective, and more convenient alternative for the emergency treatment of (Type I) allergic reactions, including anaphylaxis or what's commonly referred to as severe allergic reactions.

In addition to being a safe and effective alternative to injections, *neffy* is anticipated to have significant clinical benefit by providing patients and caregivers with an easier-to-use, needle-free device that eliminates the fear and uncertainty associated with injectable products.

REFERENCES

1. Noimark L, Wales J, Du Toit G, Pastacaldi C, Haddad D, Gardner J, Hyer W, Vance G, Townshend C, Alfaham M, Arkwright PD, Rao R, Kapoor S, Summerfield A, Warner JO, Roberts G. The use of adrenaline autoinjectors by children and teenagers. Clin Exp Allergy. 2012 Feb;42(2):284-92.
2. Asthma and Allergy Foundation of America. My Life with Food Allergy Parent Survey Report. Asthma and Allergy Foundation of America. 2019.
3. Fleming JT, Clark S, Camargo CA Jr, Rudders SA. Early treatment of food-induced anaphylaxis with epinephrine is associated with a lower risk of hospitalization. J Allergy Clin Immunol Pract. 2015 Jan-Feb;3(1):57-62.
4. Tanimoto S, Simons F, Lockey R, Lieberman P, Kaliner M, Lowenthal R. A phase 1, five-period, five-treatment, randomized crossover study of the pharmacokinetics (PK) and pharmacodynamics (PD) of epinephrine after administration of intranasal (IN) ARS-1 and intramuscular (IM) epinephrine to healthy volunteers. Journal of Allergy and Clinical Immunology. 2020;145(2):AB77
5. Westfall TC, Westfall DP. Adrenergic agonists and antagonists. In: Brunton LL, Chabner BA, Knollmann BC, editors. Goodman and Gilman's The Pharmacological Basis of Therapeutics. 12th ed. New York, NY: McGraw-Hill Companies, Inc; 2011. In. eds.

Presented at the Eastern Food Allergy and Comorbidity Conference; January 5, 2023; Palm Beach, FL